Modelling and simulation of spontaneously reported adverse events after administration of lacosamide

Rik Schoemaker[1], Armel Stockis[2]

[1]Exprimo, Mechelen, Belgium and [2]UCB Pharma, Braine l'Alleud, Belgium

Introduction

- Lacosamide (Vimpat®) is a newer anti-epileptic drug approved in the EU and US for the adjunctive treatment of adults with focal epilepsy
- Data in 193 healthy subjects (multiple dose, immediate release [IR] tablets: placebo, 400 mg and 800 mg/day, given BID) and 14 healthy subjects (single 200 mg dose, modified release [MR] form) were combined to describe the pharmacokinetics of lacosamide, using non-linear mixed-effects modelling implemented in NONMEM V7.2.0 (FOCE with interaction).
- The effect of lacosamide on the five spontaneously reported adverse events (AEs) nausea, vomiting, dizziness, oral hypoaesthesia and headache observed in the multiple dose study was modelled using a proportional odds model.
- The study objective was to predict the change in AEs incidence resulting from a MR formulation with reduced C_{max}

Methods

- PK: one-compartment model with first order absorption, diurnal effect on clearance and combined (multiplicative + additive) error
- PD: The ordered categorical data were described using proportional odds models that generate the population probabilities of a particular AE score over time using cumulative scores for Moderate and Mild+Moderate.
- With spontaneously reported AEs, consecutive scores are very highly correlated. Zingmark *et al* (1) described a Markov-type extension of the proportional odds model where the probability of obtaining a particular score depended on the preceding score.
- The parameters quantify the logit of the probability of obtaining for instance a Moderate AE given the preceding day had a Moderate, Mild or Absent AE score respectively.
- Scores of Mild+Moderate are given by parameters that shift the logit curves up from Moderate to Mild+Moderate.
- Inter-individual variability was implemented as additive distributions on the logit scale. The NONMEM parameter estimation used the Laplacian method.
- The influence of lacosamide concentrations was incorporated by using the final PK model to predict lacosamide concentrations at each corresponding AE time point using the individual empirical Bayes estimates.
- It was apparent that towards the end of the trial when lacosamide concentrations are still at their maximum, the fraction of subjects with adverse events decreases. The time-dependency was modelled by assuming an underlying slow infusion of a hypothetical substance that reduces the slope of the concentration-effect relationship. The half-life of this substance was fixed at 72 hours without inter-individual variability for all AEs because the data did not allow actual estimation of its kinetics. The 'tolerance-concentration' was scaled so that it would have a steady state value of 0.5 at 400 mg/day and of 1 at 800 mg/day. Slope reduction was implemented using an E_{max} model.

Results

- The final PK model allowed simulation of the multiple administration of the MR formulation (200 mg and 400 mg QD) in comparison with administration of the regular IR tablet (200 mg BID).
- Simulations suggest that these two administration modes cover a similar concentration range with lower peaks for the MR formulation:

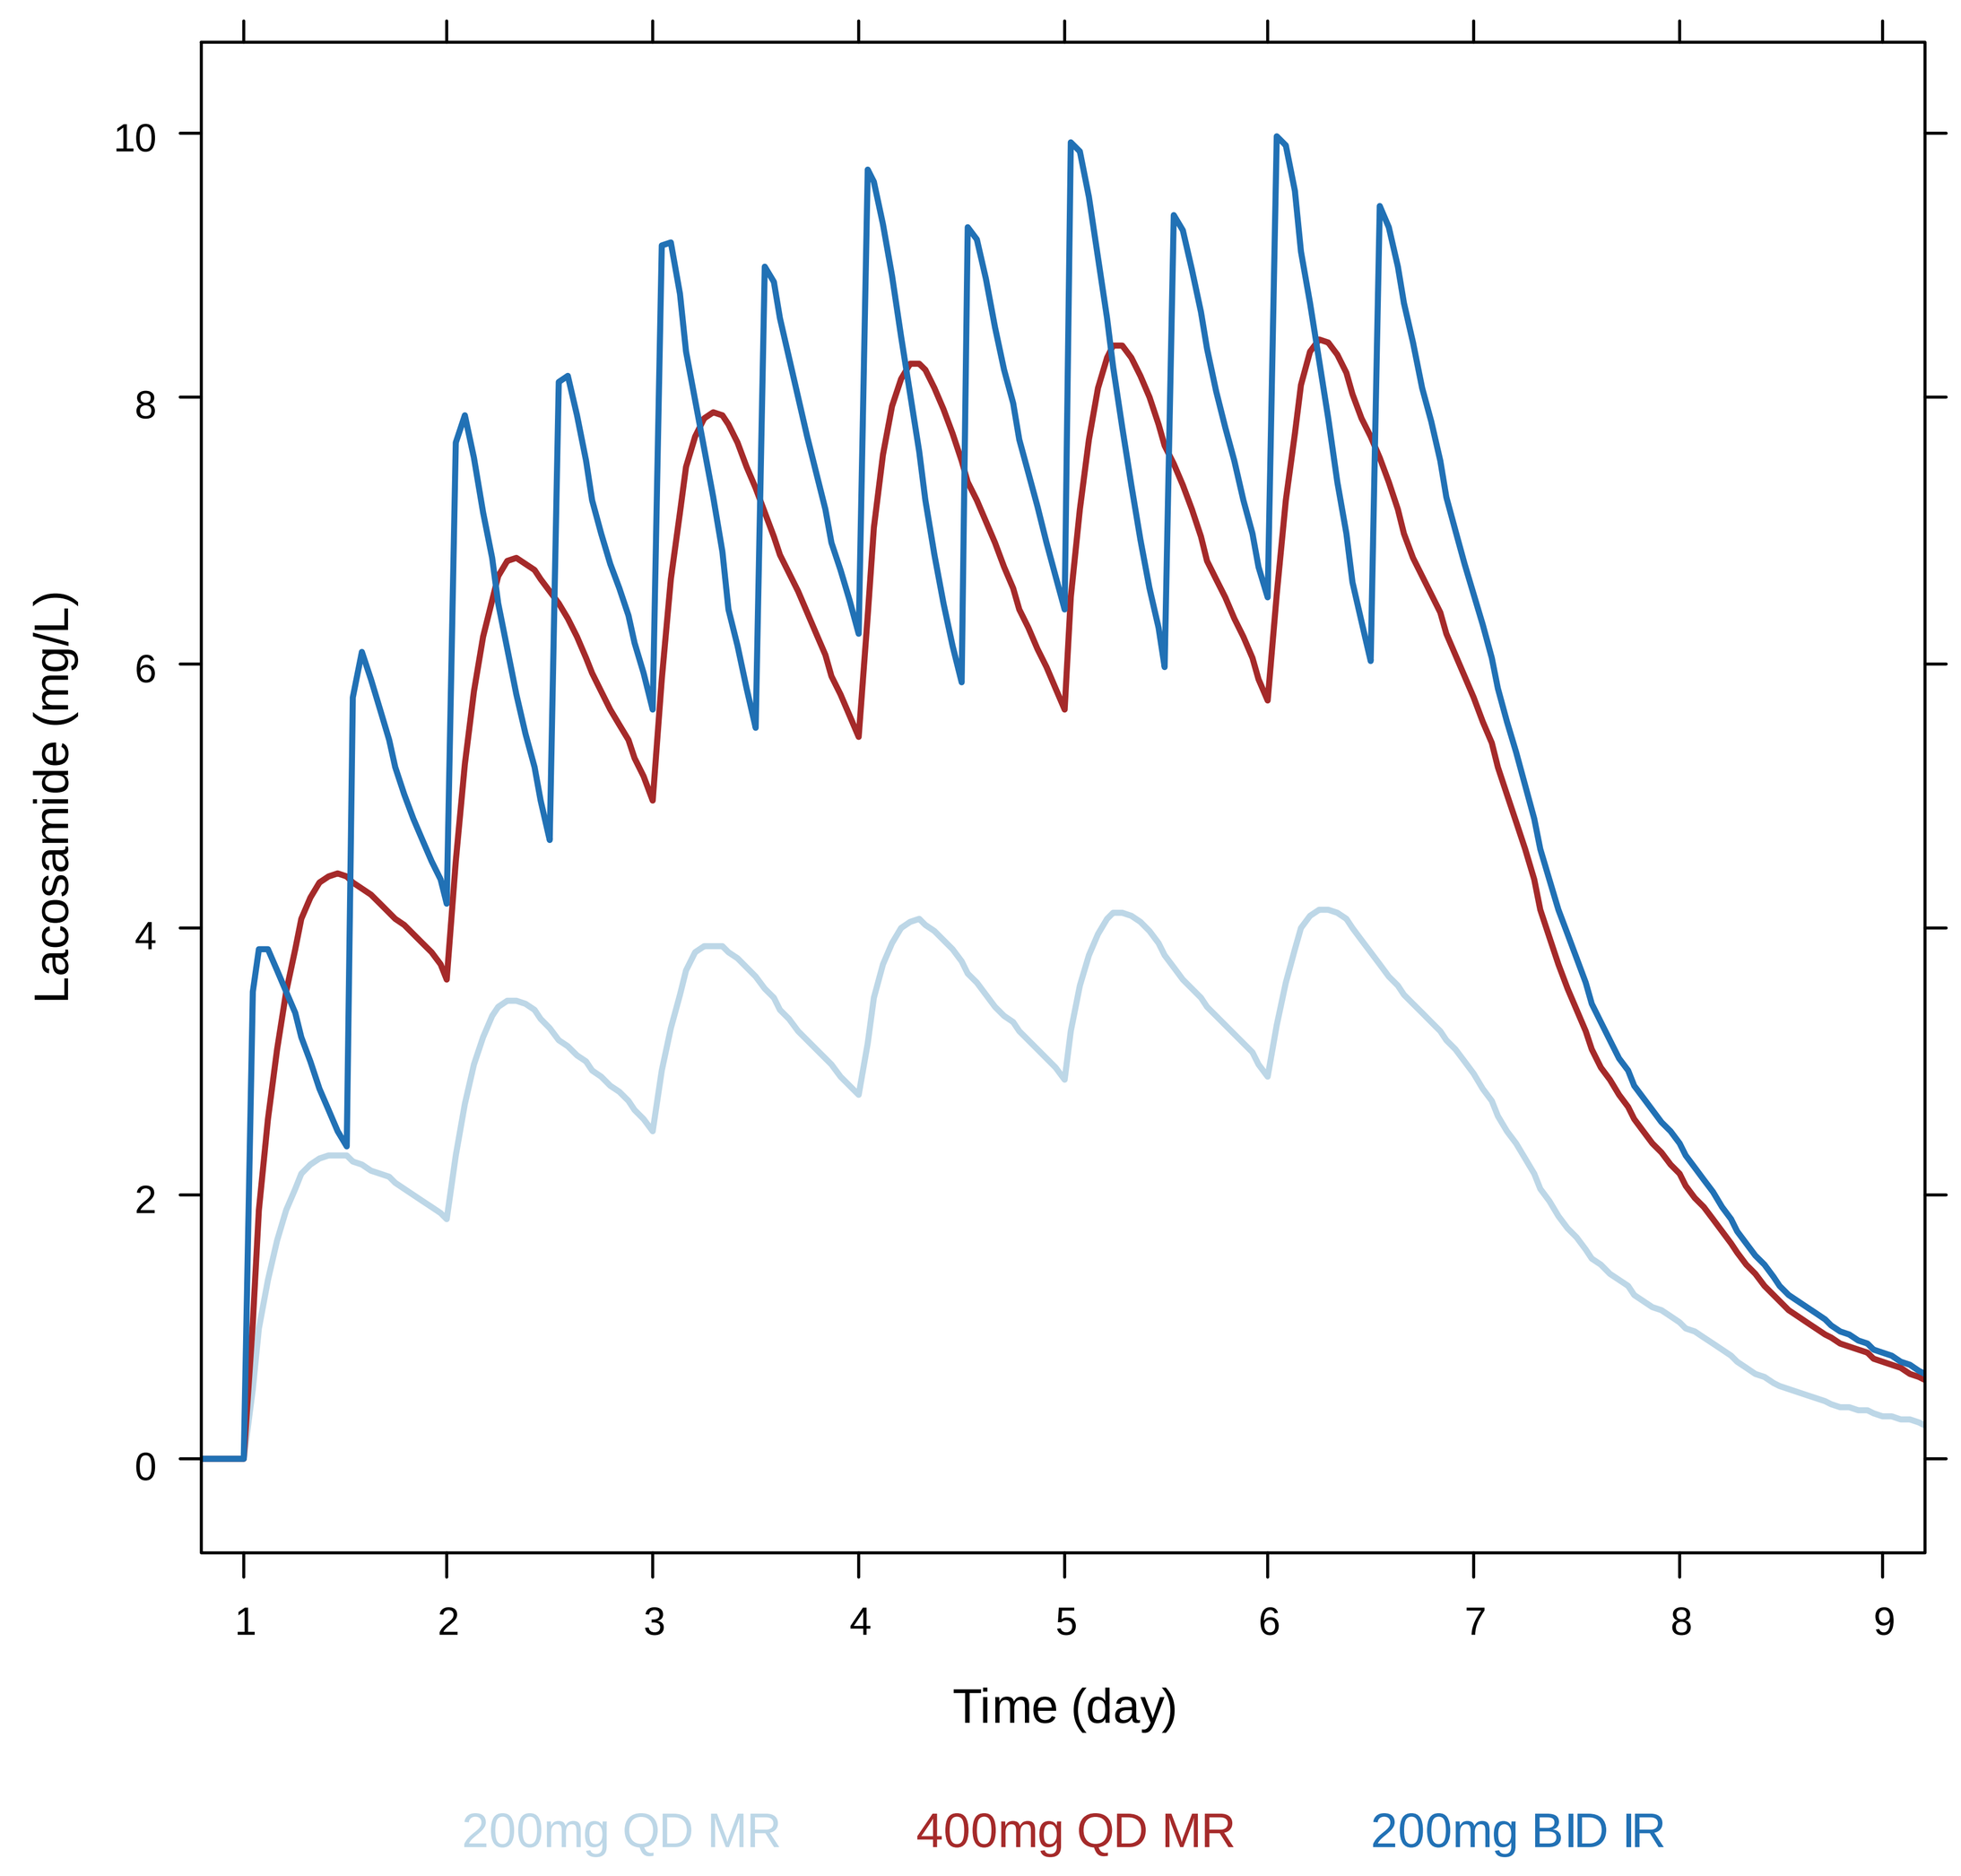
Lacosamide (mg/L)
0
2
4
6
8
10
1
2
3
4
5
6
7
8
9
Time (day)
200mg QD MR
400mg QD MR
200mg BID IR

- Visual predictive checks, illustrated below for the nausea AE, showed close correspondence between observed (red lines) and model-predicted (blue shaded areas) profiles in the multiple dose study:

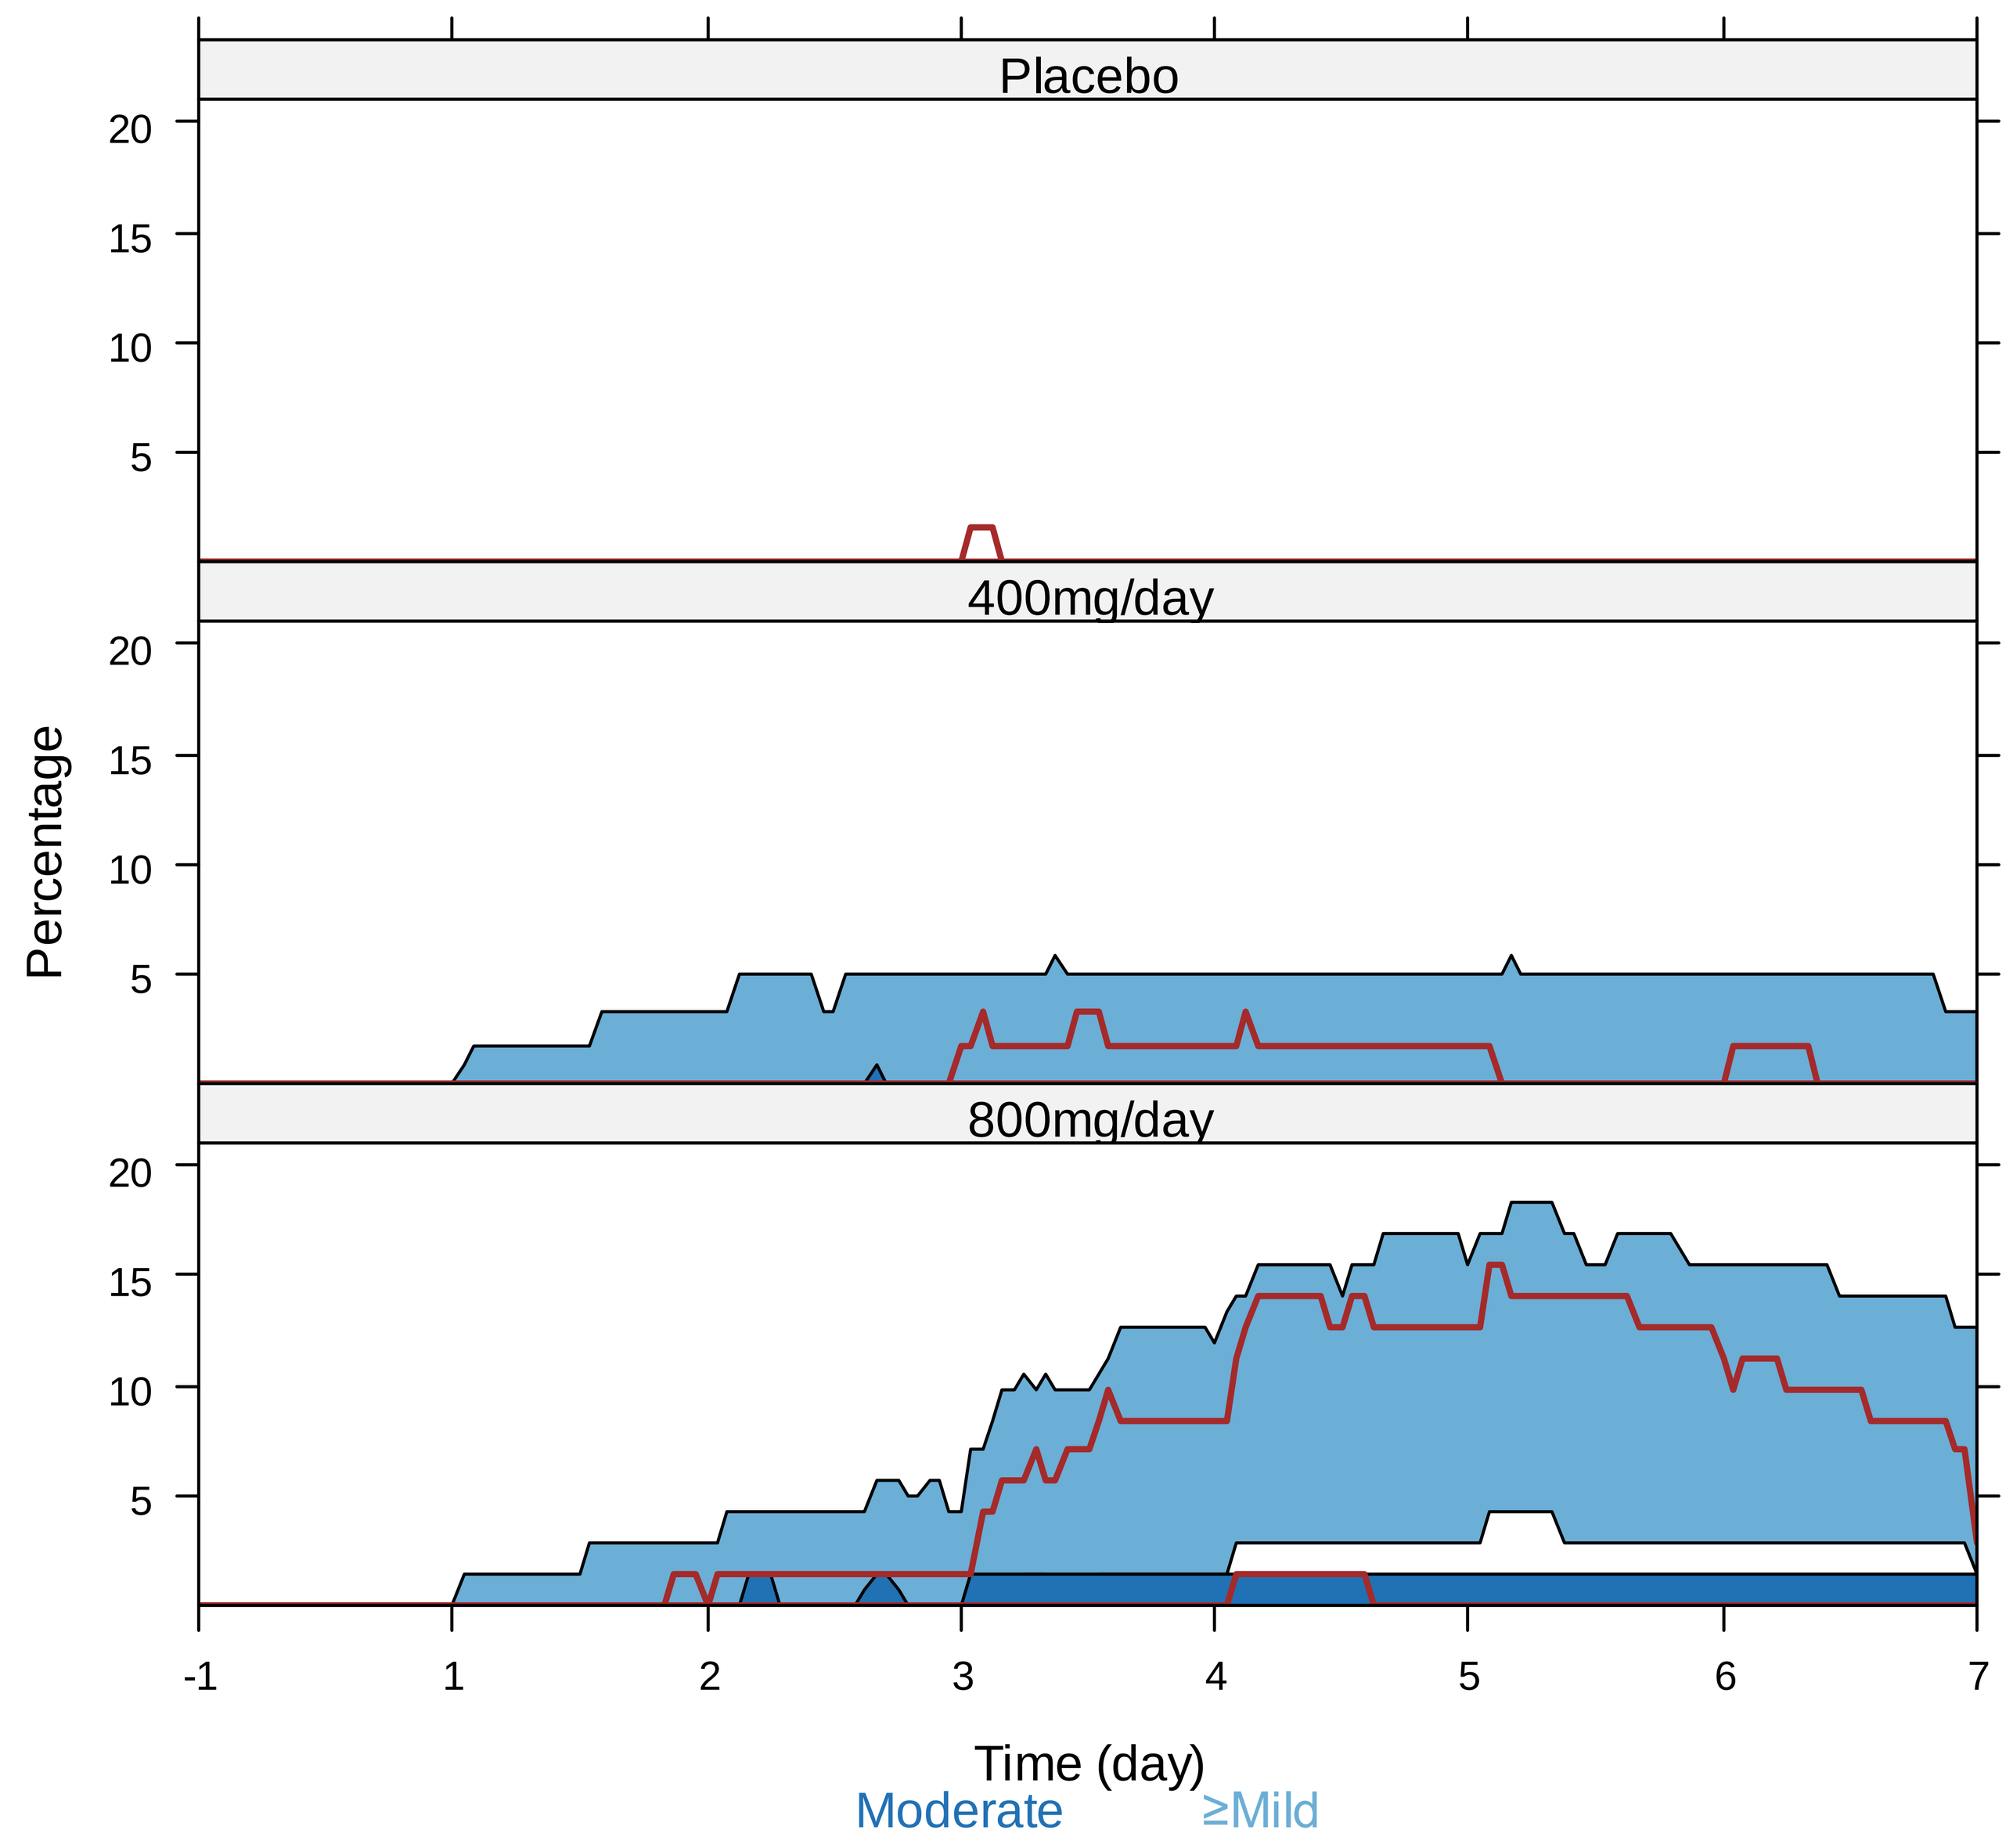
Placebo
400mg/day
800mg/day
Percentage
5
10
15
20
-1
1
2
3
4
5
6
7
Time (day)
Moderate
≥Mild

- Subsequent simulations allowed a prediction of the mean AE scores over time after IR and MR administration of lacosamide (illustrated for the nausea AE):

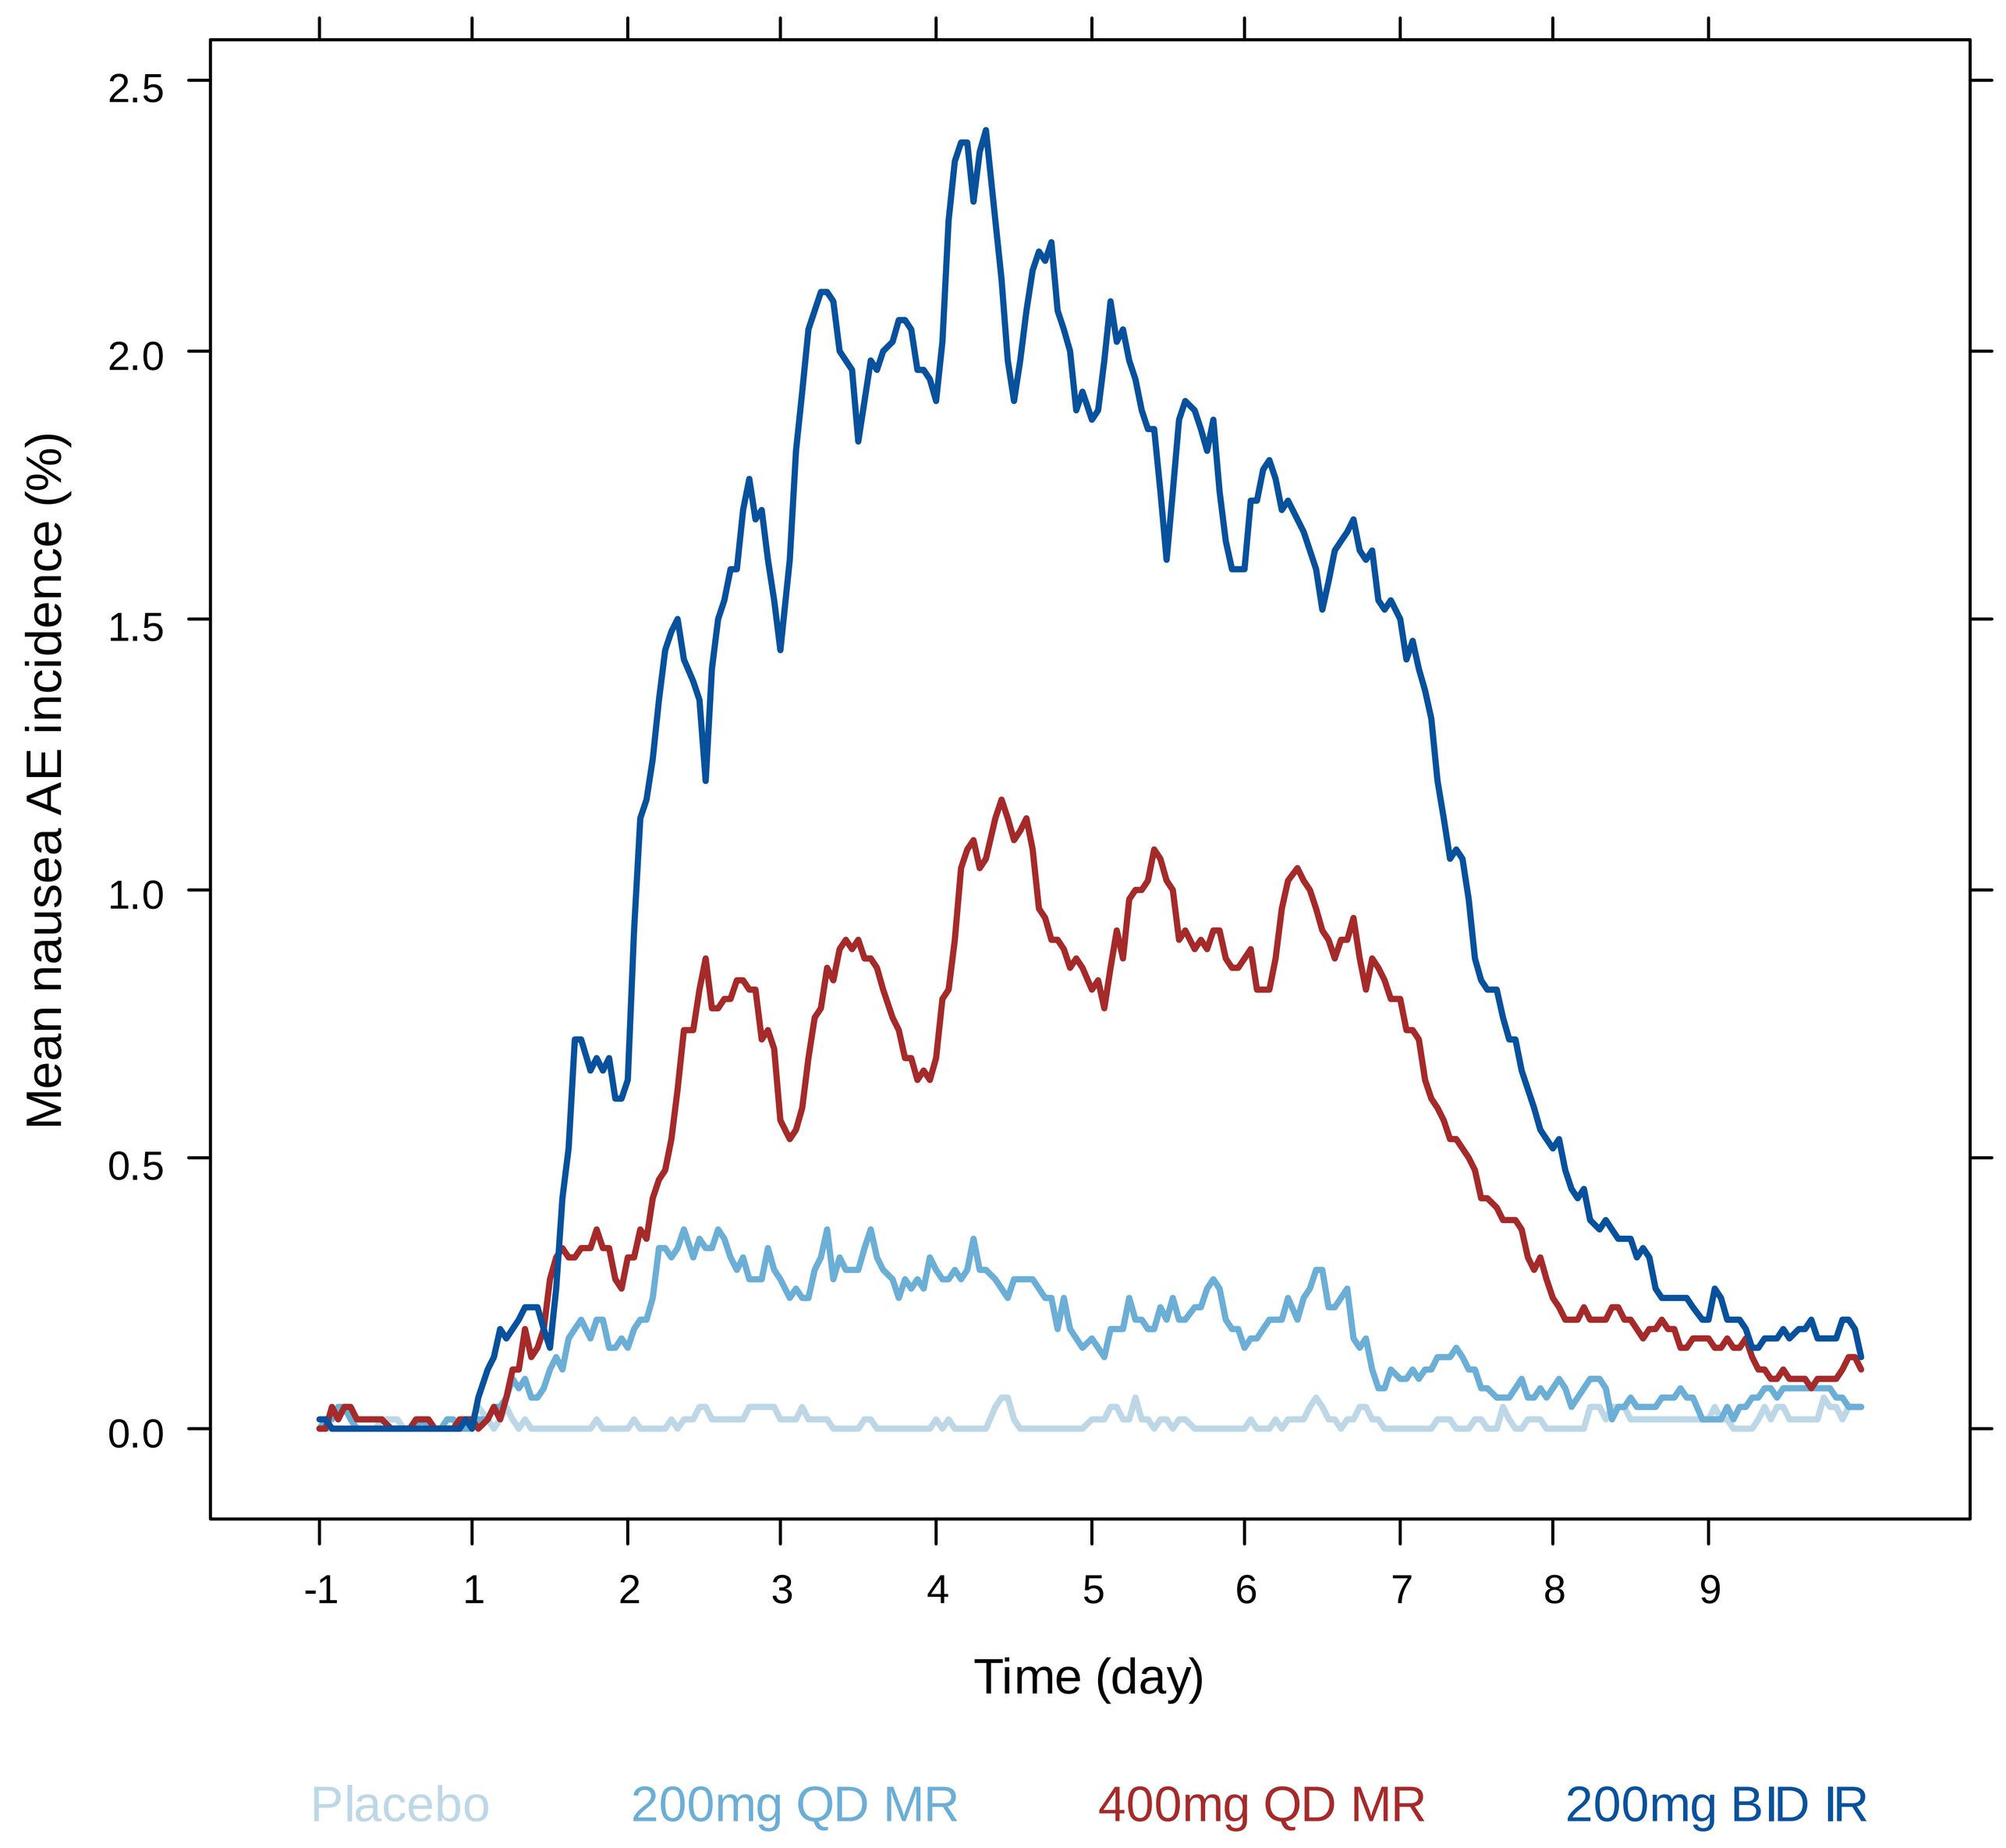
Mean nausea AE incidence (%)
0.0
0.5
1.0
1.5
2.0
2.5
-1
1
2
3
4
5
6
7
8
9
Time (day)
Placebo
200mg QD MR
400mg QD MR
200mg BID IR

For a daily 400mg lacosamide dose, simulations predicted a lower overall incidence of AEs with the MR form than with the IR tablet:

- Dizziness 20.2% vs. 25.7%
- Headache 18.7% vs. 19.5%
- Nausea 8.3% vs. 11.0%
- Oral hypoesthesia 7.3% vs. 9.9%
- Vomiting 3.0% vs. 4.8%

The difference could be explained by the difference in

- Peak concentrations (15% lower median C_{max} for MR)
- Bioavailability (7% lower for MR)

Conclusions

- Modelling and simulation of lacosamide pharmacokinetics and of the spontaneously reported AEs suggest that slowing down the absorption rate can possibly result in improved tolerability.

Reference

1) Zingmark PH, Kågedal M, Karlsson MO. Modelling a spontaneously reported side effect by use of a Markov mixed-effects model. J Pharmacokin Pharmacodyn (2005) 32: 261-81.